#71

Dual-Action Androgen Receptor Inhibitors are N-Terminal Domain Binding Androgen Receptor (AR) Antagonists and Degraders for the Treatment of AR and AR-Splice Variant-Positive Castration-Resistant Prostate Cancer

Ramesh Narayanan[1,6], Suriyan Ponnusamy[1], Yali He[2] , Dong-Jin Hwang[2], Salma Kaochar[3], Nicholas Mitsiades[3], Eva Corey[4], James B. Breitmeyer[5], Duane D. Miller[2,6], Gunnar F. Kaufmann[5]
[1] Department of Medicine, [2] Department of Pharmaceutical Sciences, University of Tennessee Health Science Center, Memphis, TN, [3] Department of Medicine, Baylor College of Medicine,
[4] Department of Urology, University of Washington, Seattle, WA, [5] Oncternal therapeutics Inc., San Diego, CA, . [6] UTHSC Center for Cancer Research

THE UNIVERSITY OF TENNESSEE HEALTH SCIENCE CENTER.

ONCTERNAL therapeutics

Introduction

- Androgen receptor (AR) is the primary therapeutic target that is expressed in over 85% of prostate cancer (PC).
- PC that relapses from hormonal treatment (castration-resistant prostate cancer (CRPC)) is aggressive and contributes to majority of PC-related deaths.
- Second-generation AR antagonists and androgen-synthesizing enzyme inhibitors have significantly extended the survival of CRPC patients.
- After a brief period of response, resistance to AR-pathway inhibitors develops by various mechanisms.
- Expression of ligand-binding domain (LBD)-null AR splice variants (AR-SVs) has been identified as one of the mechanisms by which resistance to AR-pathway inhibitors develops in CRPC. No approved drugs inhibit AR-SVs.
- **The objective of this work is to evaluate N-terminal domain-binding Dual Acting AR Inhibitors (DAARIs) in AR-SV-positive preclinical models.**

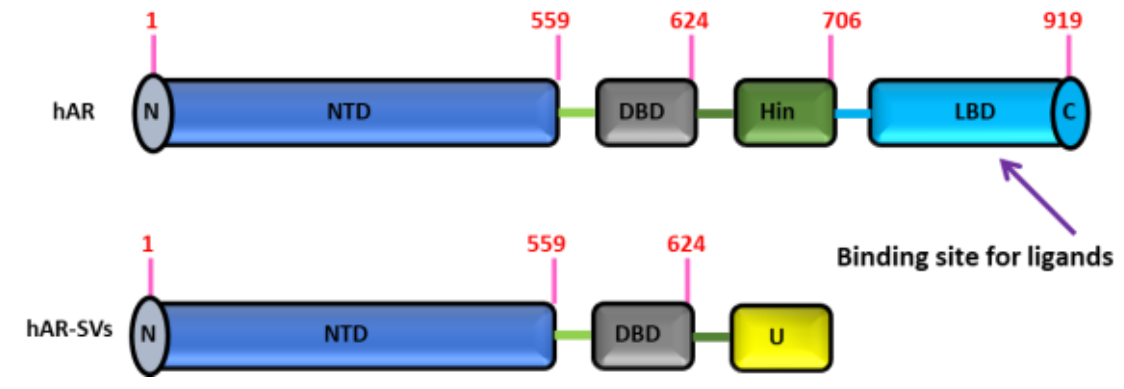
1
559
624
706
919
hAR
N
NTD
DBD
Hin
LBD
C
Binding site for ligands
1
559
624
hAR-SVs
N
NTD
DBD
U

Summary

- DAARIs ONCT-534 and ONCT-505 bind to the NTD, degrade AR & AR-SV proteins, and inhibit AR activity.
- DAARIs exhibit sustained AR inhibition in PC cells with maximum effect observed long-term treatment.
- DAARIs inhibit AR and AR-SV–positive cell line and patient-derived PC xenografts.

Conclusions: DAARIs could be promising next-generation AR-targeting drugs for the treatment of AR-SV-positive CRPCs.

Disclosure: RN, SP, DH, YH, DDM are inventors in DAARI patents. JBB and GFK are employees of Oncternal Therapeutics, Inc. (NASDAQ: ONCT), San Diego, CA and receives compensation and holds equity. RN is a consultant to ONCT. DAARI program is licensed to ONCT.

Funding: The work was funded by CA229164 grant from NCI and by ONCT.

DAARIs Degrade and Antagonize AR and AR-SV

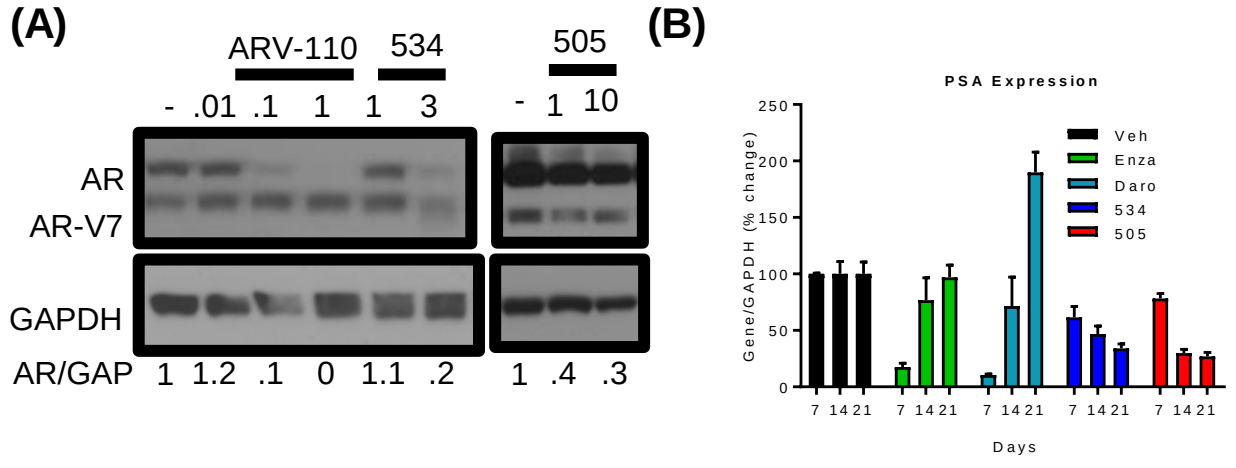
(A)
ARV-110
534
505
- .01 .1 1 1 3
- 1 10
AR
AR-V7
GAPDH
AR/GAP 1 1.2 .1 0 1.1 .2
1 .4 .3
(B)
PSA Expression
Veh
Enza
Daro
534
505
Gene/GAPDH (% change)
Days

Figure-1: **A.** DAARIs (ONCT-534 and ONCT-505) destabilize AR and AR-V7 in 22RV1 cells, while PROTAC ARV-110 destabilized only AR, but not AR-V7. **B.** DAARIs have a distinct temporal inhibition of AR function (AR-target gene FKBP5) with sustained inhibition observed in 3-Dimensional LNCaP culture treated for 21 days.

DAARIs Bind to the N-Terminal Domain of the AR

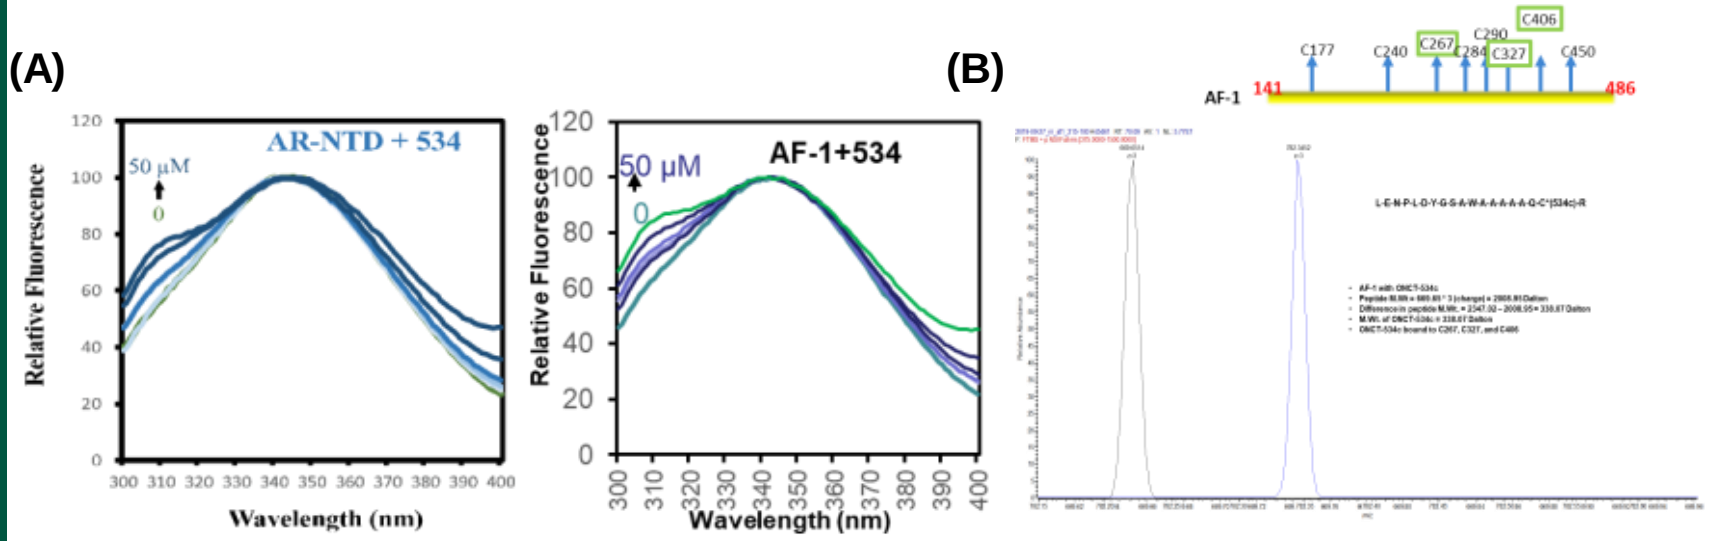
(A)
AR-NTD + 534
50 μM
0
Relative Fluorescence
Wavelength (nm)
AF-1+534
50 μM
0
Relative Fluorescence
Wavelength (nm)
(B)
AF-1
141
486
C177
C240
C267
C284
C290
C327
C406
C450

Figure-2: A. DAARI ONCT-534 binds to the N-terminal domain of the AR as shown by intrinsic fluorescence. **B**. Mass Spectrum of a covalent molecule designed from ONCT-534 confirms the binding region in the AF-1 domain. Binding amino acids are shown above the spectrum.

DAARIs Regress Enza-Resistant Tumors in CDX Models

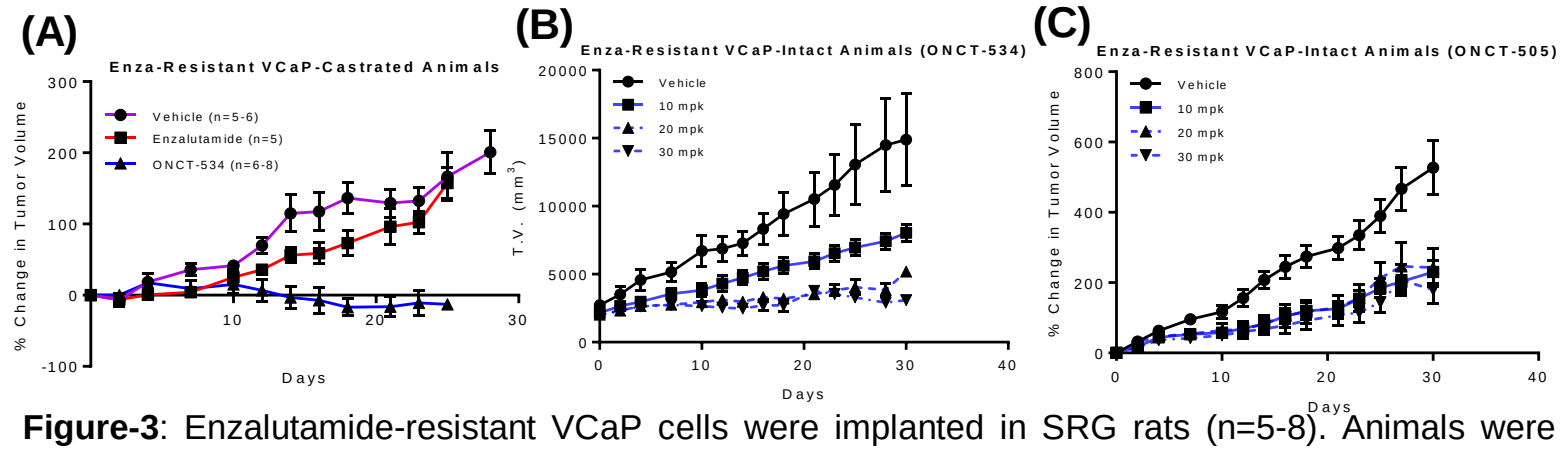
(A)
Enza-Resistant VCaP-Castrated Animals
Vehicle (n=5-6)
Enzalutamide (n=5)
ONCT-534 (n=6-8)
% Change in Tumor Volume
Days
(B)
Enza-Resistant VCaP-Intact Animals (ONCT-534)
Vehicle
10 mpk
20 mpk
30 mpk
T.V. (mm3)
Days
(C)
Enza-Resistant VCaP-Intact Animals (ONCT-505)
Vehicle
10 mpk
20 mpk
30 mpk
% Change in Tumor Volume
Days

Figure-3: Enzalutamide-resistant VCaP cells were implanted in SRG rats (n=5-8). Animals were either castrated (A) or left intact (B & C) and were treated orally with 60 mpk ONCT-534 (A), dose response of ONCT-534 (B) or ONCT-505 (C). Tumor volume and body weight were measured thrice weekly.

DAARIs Inhibit AR-SV-Positive PC Cell Line-Derived Xenograft (CDX)

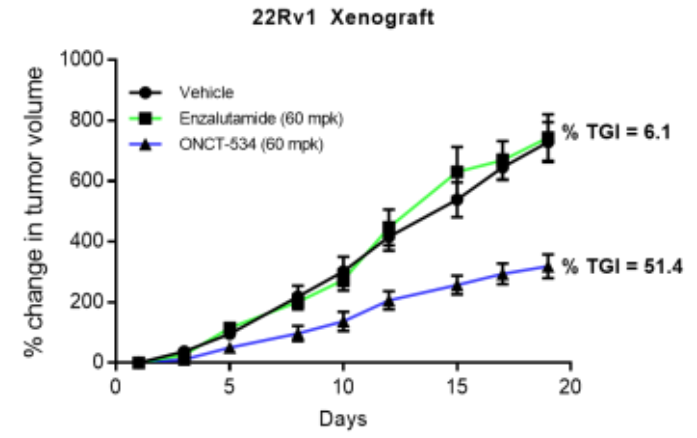
22Rv1 Xenograft
Vehicle
Enzalutamide (60 mpk)
ONCT-534 (60 mpk)
% TGI = 6.1
% TGI = 51.4
% change in tumor volume
Days

Days (p-value)	1	3	5	8	10	12	15	17	19
Enza		0.5	0.71	0.92	0.89	0.92	0.64	0.95	0.99
534		0.05	0.21	0.04	0.04	0.05	0.047	0.004	0.008
534 vs enza			0.04	0.07	0.07	0.03	0.011	0.003	0.006

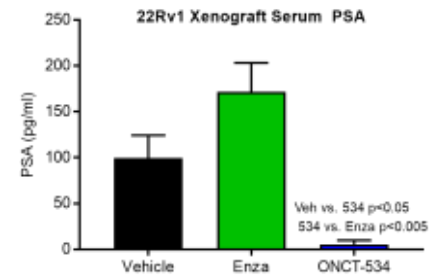
22Rv1 Xenograft Serum PSA
PSA (pg/ml)
Veh vs. 534 p<0.05
534 vs. Enza p<0.005
Vehicle
Enza
ONCT-534

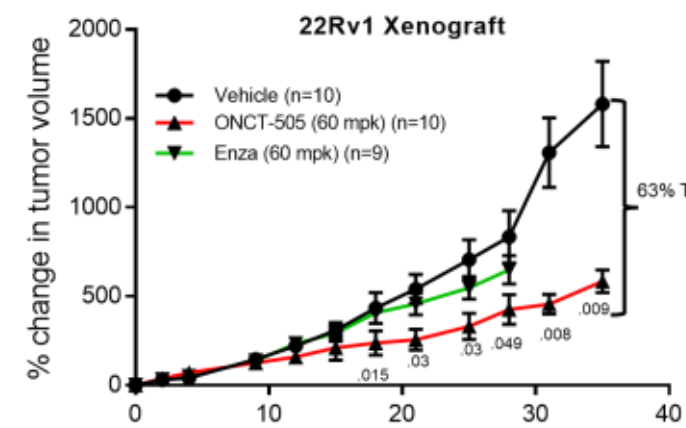
22Rv1 Xenograft
Vehicle (n=10)
ONCT-505 (60 mpk) (n=10)
Enza (60 mpk) (n=9)
63% TGI
% change in tumor volume
Days

Mice attaining euthanasia criteria before 28 days:
Vehicle = 5/10
ONCT-505 = 2/10
Enza = 5/9

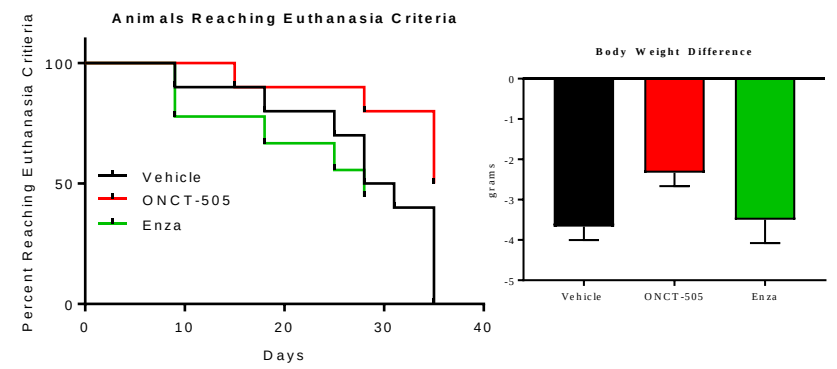
Animals Reaching Euthanasia Criteria
Vehicle
ONCT-505
Enza
Percent Reaching Euthanasia Critieria
Days
Body Weight Difference
grams
Vehicle
ONCT-505
Enza

Figure-4: A. 22Rv1 cells were implanted in SRG immunocompromised rats. Once the tumors are palpable, the animals were castrated, and the tumors were allowed to regrow as CRPC. Animals (n=7-10) were randomized and treated orally. Tumor volume was measured thrice weekly. Serum PSA was measured in serum collected at the end of the study. **Right**. 22Rv1 cells were implanted in NSG mice and a xenograft study was performed as indicated for the left panel.

DAARIs Inhibit AR-SV-Positive Patient-Derived Xenograft (PDX)

Details of LuCaP 86.2 PDX cells
- obtained from bladder metastasis
- expresses AR and AR v567es splice variant.
- No response to AR-targeting agents, such as Enza or Abi
- Response to docetaxel.

REFERENCES:
- Ngyun HM., Prostate, 77, 654-671, 2017
- Zhang x., Plos One., 6, e27970, 2011
- Lam HM., Clin Can Res. 23(9), 2301-12, 2017
- Winters B., AntiCancer Res., 37, 3385-96, 2017

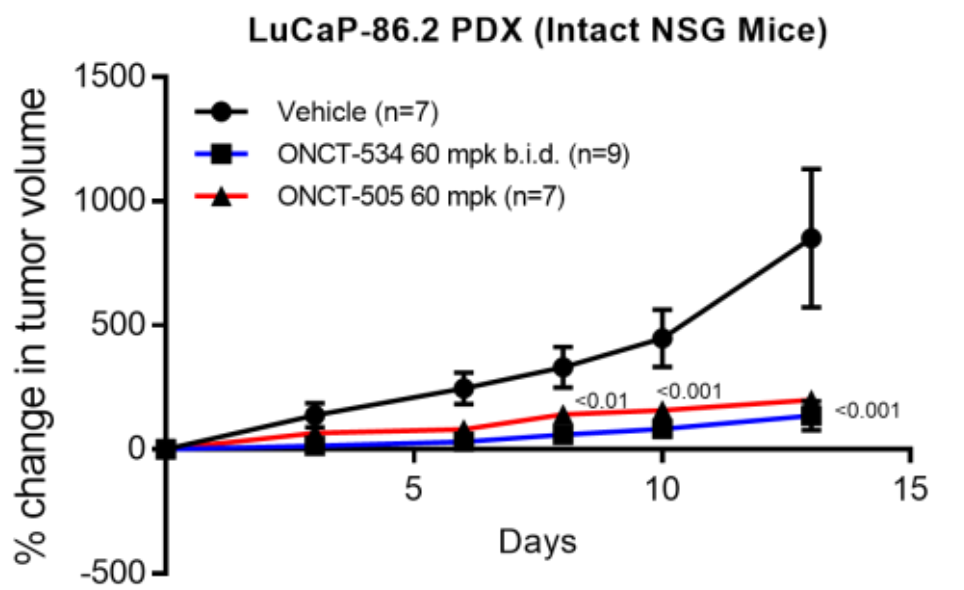
LuCaP-86.2 PDX (Intact NSG Mice)
Vehicle (n=7)
ONCT-534 60 mpk b.i.d. (n=9)
ONCT-505 60 mpk (n=7)
<0.01
<0.001
<0.001
% change in tumor volume
Days

Figure-5: LuCaP 86.2 PDX tumor pieces were implanted into intact NSG mice. Once the tumors reached 100-300 mm^3, the animals were randomized and treated orally.